Anti-inflammatory and Immunomodulatory Activities of Parnaparin Sodium (CB-01-05)

C. Rosette[1], L. Moro[2], G. Celasco[2], R. Bozzella[2], M. Ajani[2], M. Gerloni[1]
[1]Cosmo Bioscience, San Diego, United States. [2] Cosmo Pharmaceuticals, Lainate, Italy

1. Introduction

Phase II clinical trials with oral low molecular weight heparin, parnaparin sodium (CB-01-05) assembled with MMX® technology, showed promising efficacy in improving ulcerative colitis symptoms. The mechanisms of action underlying this therapeutic effect have not yet been elucidated. Since IBD is characterized by inflammation and immune disregulation, we investigated *in vitro* whether CB-01-05 exhibitis anti-inflammatory and immunomodulatory activities to account for the positive effects seen in the clinical trials.

2. Conclusions

- Sodium parnaparin inhibited TNF-a and IL-1b-induced synthesis and secretion of the chemokine IL-8 from human intestinal epithelial cell line.
- Sodium parnaparin inhibited secretion of IL6 from human monocytes.
- Sodium parnaparin substantially inhibited cytokine secretion from activated CD4+ T cells: IFN-γ, IL17, TNF-α, IL6, IL-2, IL-5 and IL-10.
- Sodium parnaparin effectively prevented the differentiation of naïve CD4+ T cells into cells characteristic of mature Th1 and Th2 cells.

3. Relevance

Reduction of synthesis of several cytokines that are known to be overexpressed in inflamed colonic mucosa of IBD patients is useful to control the inflammatory response in this chronic disease. It is anticipated that the delivery of sodium parnaparin-MMX® into the colon could antagonize the local inflammatory cytokine effects, without affecting the functions of these cytokines in the periphery.

Acknowledgments

We thank Dr. R. Bacchetta for the L cells and technical guidance, K. Fontes, F.B.Laysa, M. Mascarenas, and S.Patkar for laboratory assistance.

For further information

Please contact Cari Rosette: crosette@cosmobioscience.com.
More information on this and related projects can be obtained at *www.cosmopharmaceuticals.com.*

4. Activity on Epithelial Cells

Effect of parnaparin on intestinal epithelial cell function

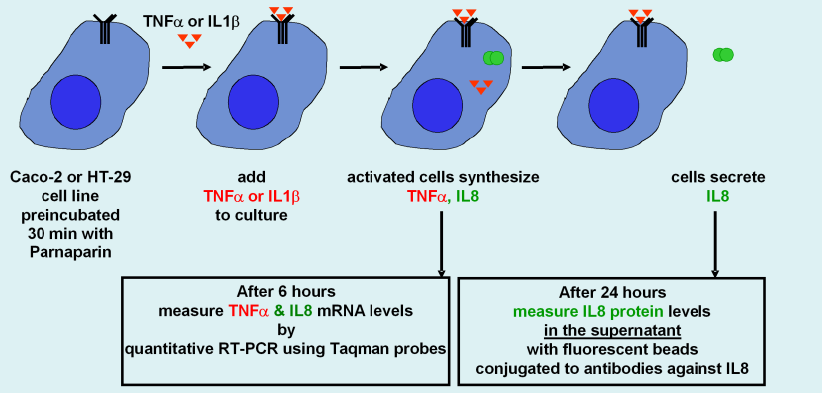
TNFα or IL1β
Caco-2 or HT-29 cell line preincubated 30 min with Parnaparin
add TNFα or IL1β to culture
activated cells synthesize TNFα, IL8
cells secrete IL8
After 6 hours measure TNFα & IL8 mRNA levels by quantitative RT-PCR using Taqman probes
After 24 hours measure IL8 protein levels in the supernatant with fluorescent beads conjugated to antibodies against IL8

Parnaparin inhibits TNFα-induced IL8 mRNA expression and IL8 protein secretion from Caco-2 cells

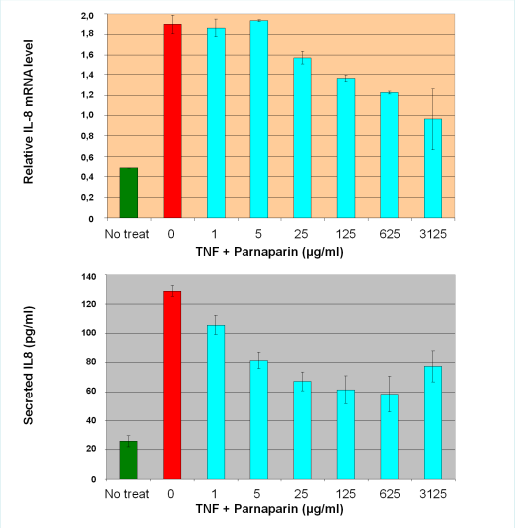
Relative IL-8 mRNA level
No treat
0
1
5
25
125
625
3125
TNF + Parnaparin (µg/ml)
Secreted IL8 (pg/ml)

IL8 mRNA
6hr TNFα stimulation

Secreted IL8 protein
24hr TNFα stimulation

Parnaparin inhibits TNFα-induced IL8 secretion from Caco-2

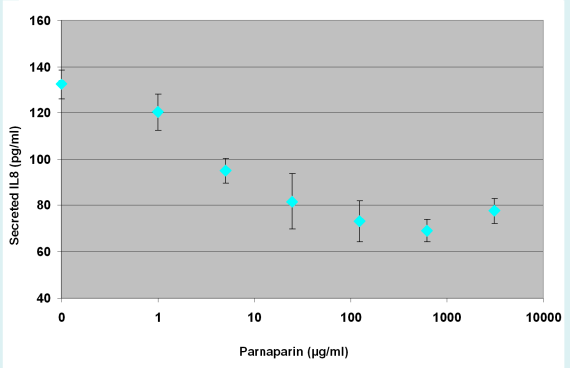
Secreted IL8 (pg/ml)
Parnaparin (µg/ml)

N = 4 experiments

Parnaparin inhibits IL1β-induced IL8 secretion from Caco-2

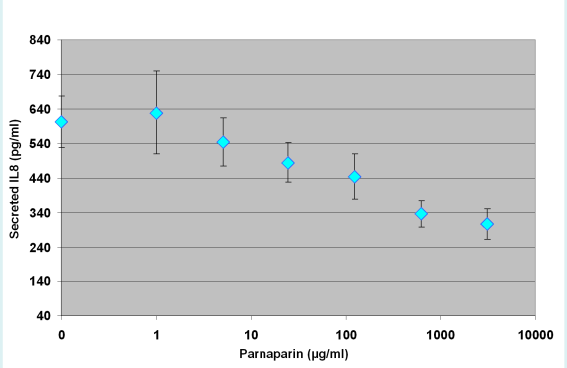
Secreted IL8 (pg/ml)
Parnaparin (µg/ml)

N = 6 experiments

5. Activity on Monocytes

Parnaparin inhibits IL6 from Monocytes

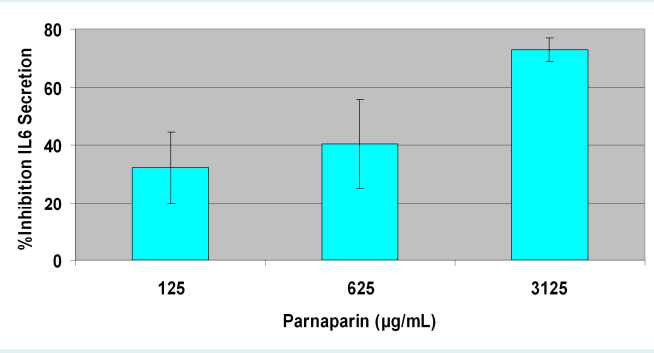
%Inhibition IL6 Secretion
125
625
3125
Parnaparin (µg/mL)

N = 3 experiments

6. Activity on CD4 T Cells

Inhibition of cytokines from CD4+ cells

Ficoll separated PBMC

Purify **CD4+** cells by magnetic beads

↓

Coat plate anti-CD3/CD28 + anti-IgG

↓

Plate 0.5 to 1x10[6] cells per well

↓

Add parnaparin, heparin or dexamethasone

↓ 24 - 48 hr / 37°C / 5%CO_2

Harvest supernants

↓

Incubate with beads to capture cytokines/chemokines

↓

Analyze fluorescence intensity by flow cytometry

Parnaparin inhibits secretion of multiple cytokines

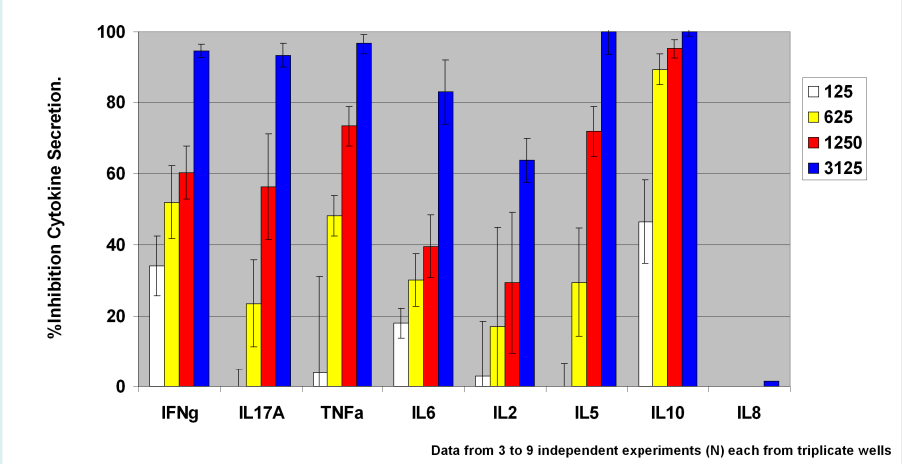
%Inhibition Cytokine Secretion.
IFNg
IL17A
TNFa
IL6
IL2
IL5
IL10
IL8
125
625
1250
3125
Data from 3 to 9 independent experiments (N) each from triplicate wells

7. Activity on CD4 Polarization

Effect of Parnaparin on Th1/Th2 differentiation *in vitro*

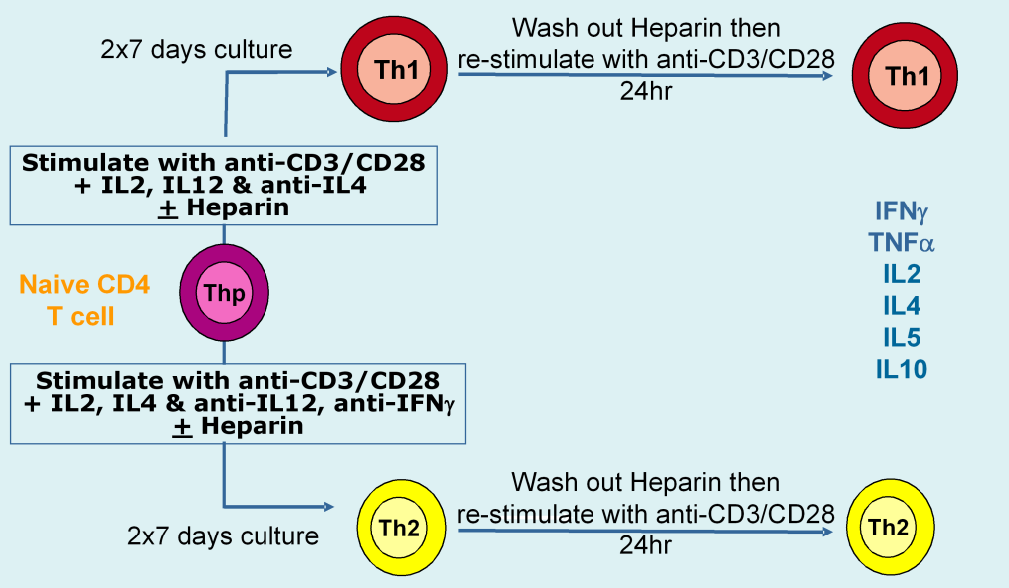
2x7 days culture
Th1
Wash out Heparin then
re-stimulate with anti-CD3/CD28
24hr
Th1
Stimulate with anti-CD3/CD28
+ IL2, IL12 & anti-IL4
± Heparin
Naive CD4
T cell
Thp
Stimulate with anti-CD3/CD28
+ IL2, IL4 & anti-IL12, anti-IFNγ
± Heparin
2x7 days culture
Th2
Wash out Heparin then
re-stimulate with anti-CD3/CD28
24hr
Th2
IFNγ
TNFα
IL2
IL4
IL5
IL10

Effect of Parnaparin on Th1/Th2 differentiation *in vitro*

Parnaparin inhibits expression of Th1 cytokines

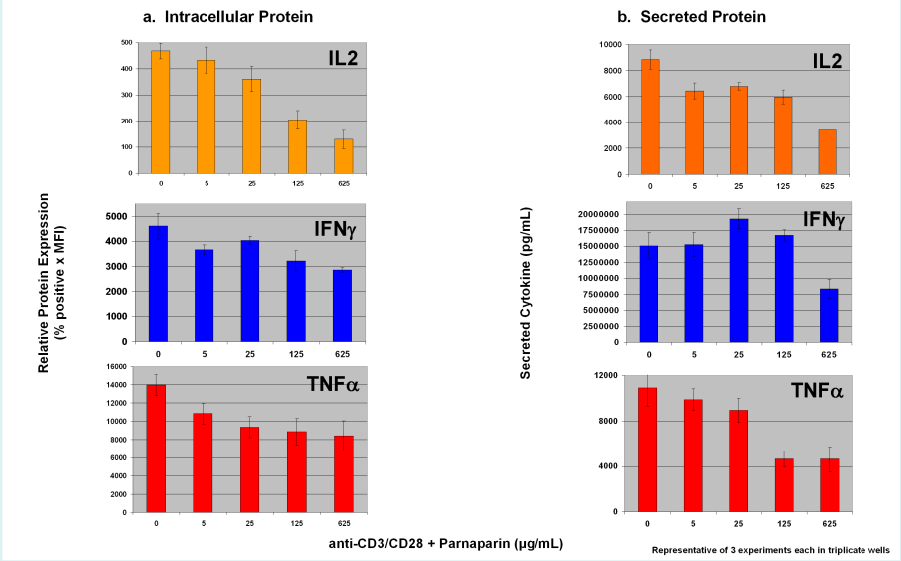
a. Intracellular Protein
b. Secreted Protein
IL2
IFNγ
TNFα
Relative Protein Expression (% positive x MFI)
Secreted Cytokine (pg/mL)
anti-CD3/CD28 + Parnaparin (µg/mL)
Representative of 3 experiments each in triplicate wells

Parnaparin inhibits CD4+ T Cell Polarization

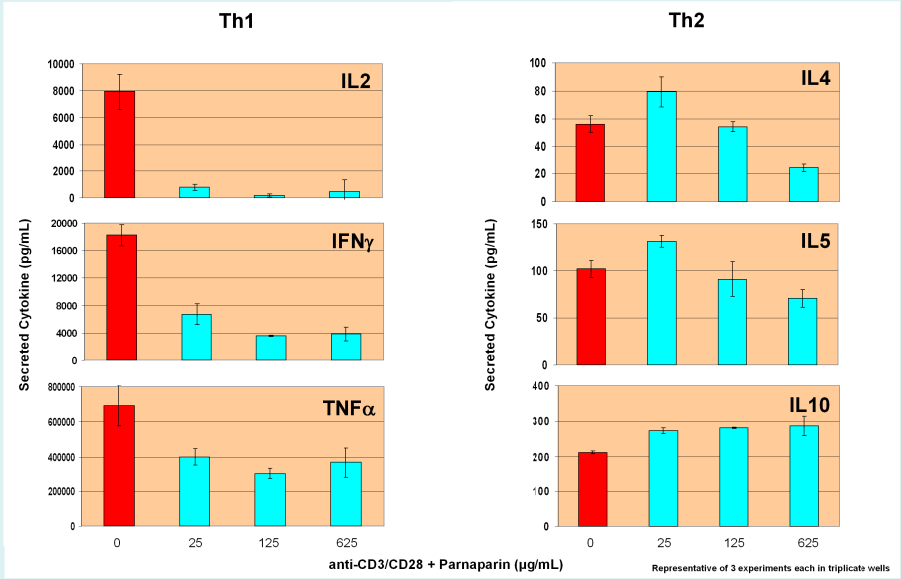
Th1
Th2
IL2
IFNγ
TNFα
IL4
IL5
IL10
Secreted Cytokine (pg/mL)
0
25
125
625
anti-CD3/CD28 + Parnaparin (µg/mL)
Representative of 3 experiments each in triplicate wells

DDW 2009 - W1616